An Inter-Laboratory Analysis of Nortriptyline CYP Interactions *In Vitro* Confirms Merits of fu_{inc} Considerations

Philip Roberts[1], **Robert J. Riley**[2], **Ruth Swan**[1], **Dean Slee**[2] and **S. Michelle Whittaker**[1], (1) School of Pharmacy and Pharmaceutical Science, University of Central Lancashire, Preston, PR1 2HE, United Kingdom, (2) Department of Physical & Metabolic Science, AstraZeneca R&D Charnwood, Bakewell Rd, Loughborough, United Kingdom

Introduction

In vitro test systems are used routinely by the pharmaceutical industry for high throughput analysis of the ADME properties of new chemical entities (NCEs). The use of rapid cytochrome P450 (CYP) inhibition screens has proved to be useful in providing information of potential drug-drug interactions. The metabolising system used may vary, but the most common systems utilise either human liver microsomes (HLM) or heterologous expression systems, with good correlation between the two systems being reported (1). However, it has also been shown that variables such as non-specific binding in these systems can influence the results obtained and hence conclusions drawn from such analyses (2). Here we report *in vitro* interactions of nortriptyline with recombinant human CYP enzymes *in vitro*, which are in contrast to some previously reported work.

Methods

Two separate *in vitro* systems were used to measure the interaction of nortriptyline with three human CYP isoforms (CYP2C19, CYP2D6 and CYP3A4 Bactosomes, Cypex).

System 1: Nortriptyline was incubated in 96 well plates with separate CYP isoforms in the presence of an NADPH regenerating system. CYP activity was monitored using the following fluorogenic substrates: CYP2C19 – CEC; CYP2D6 – AMMC; CYP3A4 - BFC. IC_{50} values were calculated and compared to those of control inhibitors (CYP2C19 – Tranycylpromine; CYP2D6 – Quinidine; CYP3A4 - Ketoconazole).

System 2: Nortriptyline was incubated in 96 well plates with a mixture of CYP isoforms in the presence of NADPH. CYP activity was monitored using the following substrates as an *in vitro* cocktail: CYP2C19 – Mephenytoin: CYP2D6 – Bufuralol: CYP3A4 - Midazolam; using LC/MS/MS detection as previously reported (3). IC_{50} values were calculated and compared to those of control inhibitors (as above).

Results

Nortriptyline was found to inhibit all of 3 CYP isoforms investigated (Fig 1). The data in Table 1 show the calculated IC_{50} values for the different incubation systems utilised, which were in good excellent agreement. Nortriptyline, in both instances, inhibited all three CYP isoforms tested to a similar degree.

Discussion

The data presented here from two laboratories indicate that nortriptyline interacts with all three CYP isoforms investigated. This contrasts with previously published data, where only an interaction with CYP2D6 was observed (4).

Furthermore, the current data concur with other reports, which suggest a role for CYP isoforms other than CYP2D6 in the metabolism of nortriptyline (5).

It is suggested that factors such as non-specific binding resulting from the high protein content of incubations adopted by some laboratories may result in false negative results for lipophilic drugs such as nortriptyline (logP = 3.4). Indeed, concentrations of both CYP and assay protein was found to be much higher in some previous studies when compared to the work described here (Table 2), although in both instances substrate concentrations used were ≤ Km for the individual CYP isoforms.

These findings emphasise further that attention to experimental design including the incubation conditions used is fundamental to enabling satisfactory conclusions on drug-CYP interactions for NCEs to be drawn.

Fig 1. Inhibition of CYP activity by nortriptyline for A) CYP 2C19; B) CYP 2D6 and C) CYP 3A4 using SYSTEM 1 (values are average of n=3 incubations)

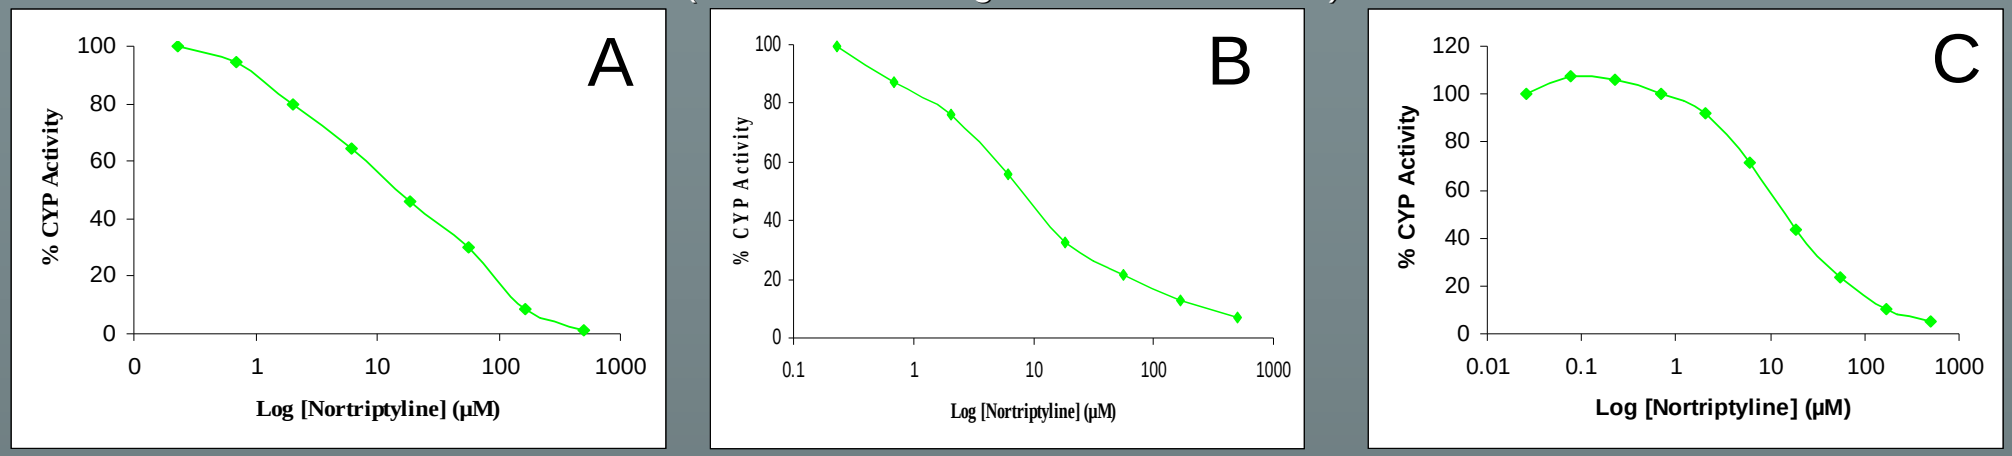
A
% CYP Activity
Log [Nortriptyline] (µM)
B
% CYP Activity
Log [Nortriptyline] (µM)
C
% CYP Activity
Log [Nortriptyline] (µM)

Table 1. IC_{50} values for Nortriptyline and Control Inhibitors in two *in vitro* test systems. Data are mean ± SD (N = 3) or from individual experiments.

CYP studied	IC_{50} (µM)			
	SYSTEM 1		SYSTEM 2	
	Control	Nortriptyline	Control	Nortriptyline
CYP2C19	3.4, 2.8	8, 9	2.9, 4.6	4 ± 2
CYP2D6	0.07 ± 0.02	11.6 ± 3.4	0.01, 0.03	5 ± 2
CYP3A4	0.03 ± 0.01	21 ± 9	0.003, 0.003	9 ± 1

Table 2. Comparison of concentrations of CYP and protein in incubations conducted in this study (SYSTEM 1) and in previous work, where no inhibition was reported (4).

	µg protein/ml	pmol CYP/ml
Current	2	4
Previous	1000 (microsomes)	200 (cDNA expressed CYP)

References

1. McGinnity, D.F. and Riley, R.J. (2001), 29, 135-139.
2. Austin, R.P., Barton, P., Cockroft, S.L., Wenlock, M.C. and Riley, R.J. (2002) Drug Met. Dispos., 30, 1497-1503.
3. Weaver, R., Graham, K.S., Beattie, I.G. and Riley, R.J. (2003), Drug Met. Dipos., 31, 955-966.
4. Shin, J., Park, J., Kim, M., Shon, J., Yoon, Y. Cha, I, Lee, S., Oh, S., Kim, S. and Flockhart, D.A.. (2002), Drug Met Dispos. 30, 1102-1107.
5. von Bahr, C., Steiner, E., Koike, Y. and Gabrielsson, J. (1998) Clin. Pharm. Ther; 64, 18-26.